The Impact of Hepatitis C Viral Cure on Progression of Renal Disease

Carla V Rodriguez[1]; Kevin B Rubenstein[1]; Jean Marie Arduino[2]; Jin-Wen Hsu[3]; Haihong Hu[1]; Rong Wei[3]; Michael Horberg[1]; Stephen Derose[3]; Sara Tartof[3]

[1]Mid-Atlantic Permanente Research Institute, Kaiser Permanente Mid-Atlantic States (KPMAS), Rockville, MD, USA ; [2]Merck & Co., Inc., Kenilworth, NJ, USA;
[3]Department of Research & Evaluation, Kaiser Permanente Southern California (KPSC), Pasadena, CA, USA

MERCK

Kaiser Permanente Research

Introduction

- Chronic hepatitis C virus (HCV) infection is associated with the development of chronic kidney disease (CKD)
- The impact of HCV cure on renal progression is poorly understood
- We compared differences in the change in estimated glomerular filtration rate (eGFR) over time and time to end-stage renal disease (ESRD) by cure status. Cure was defined as sustained viral response at 24 weeks (SVR-24) after interferon (IFN)-based therapy

Methods

- ***Study Population:*** KPSC or KPMAS patients ≥18 years of age with incident HCV diagnosed from 1/1/2004 to 12/31/2014 **and** who had completed at least 4 weeks of IFN-based therapy (including boceprevir or telaprevir). Chronic HCV was defined by at least one of: positive HCV RNA, detectable HCV genotype, ≥2 refills of anti-HCV drugs within 1 year, or positive HCV antibody test plus ≥1 HCV-coded visit
- ***Definition of SVR (exposure):*** Assessed at ≥24 weeks after termination of last therapy during the study period
- ***Definition of Baseline*****:** The year prior to the start of therapy
- ***Outcomes of Interest:*** a) Change in posttreatment eGFR (as per CKD-EPI definition); b) Time to 25% change in eGFR; c) Time to ESRD, defined as eGFR <15 mL/min/1.73 m^2 at least 3 months apart, dialysis, or kidney transplant approval
- ***Analytic Approach:*** Generalized estimating equations (GEE) to estimate effect of SVR on change in eGFR over time. Cox proportional hazards models to estimate time to ESRD and 25% change in eGFR from baseline. All models adjusted for age, sex, eGFR, HCV genotype, HCV viral load, cirrhosis, diabetes, HIV, HBV, and race at baseline
- ***Follow-up Time*****:** Censored at the earliest of death, disenrollment from KP health plan, dialysis, liver or kidney transplant, or 12/31/2014. The ESRD model was censored on death, disenrollment, or 12/31/2014

Funding: Study was funded by Merck & Co., Inc.
Conflicts of interest: JM Arduino is an employee and stockholder of Merck & Co., Inc.

Copyright © 2016 Merck Sharp & Dohme Corp., a subsidiary of **Merck & Co., Inc.** All rights reserved.

Results

Figure 1. The cohort

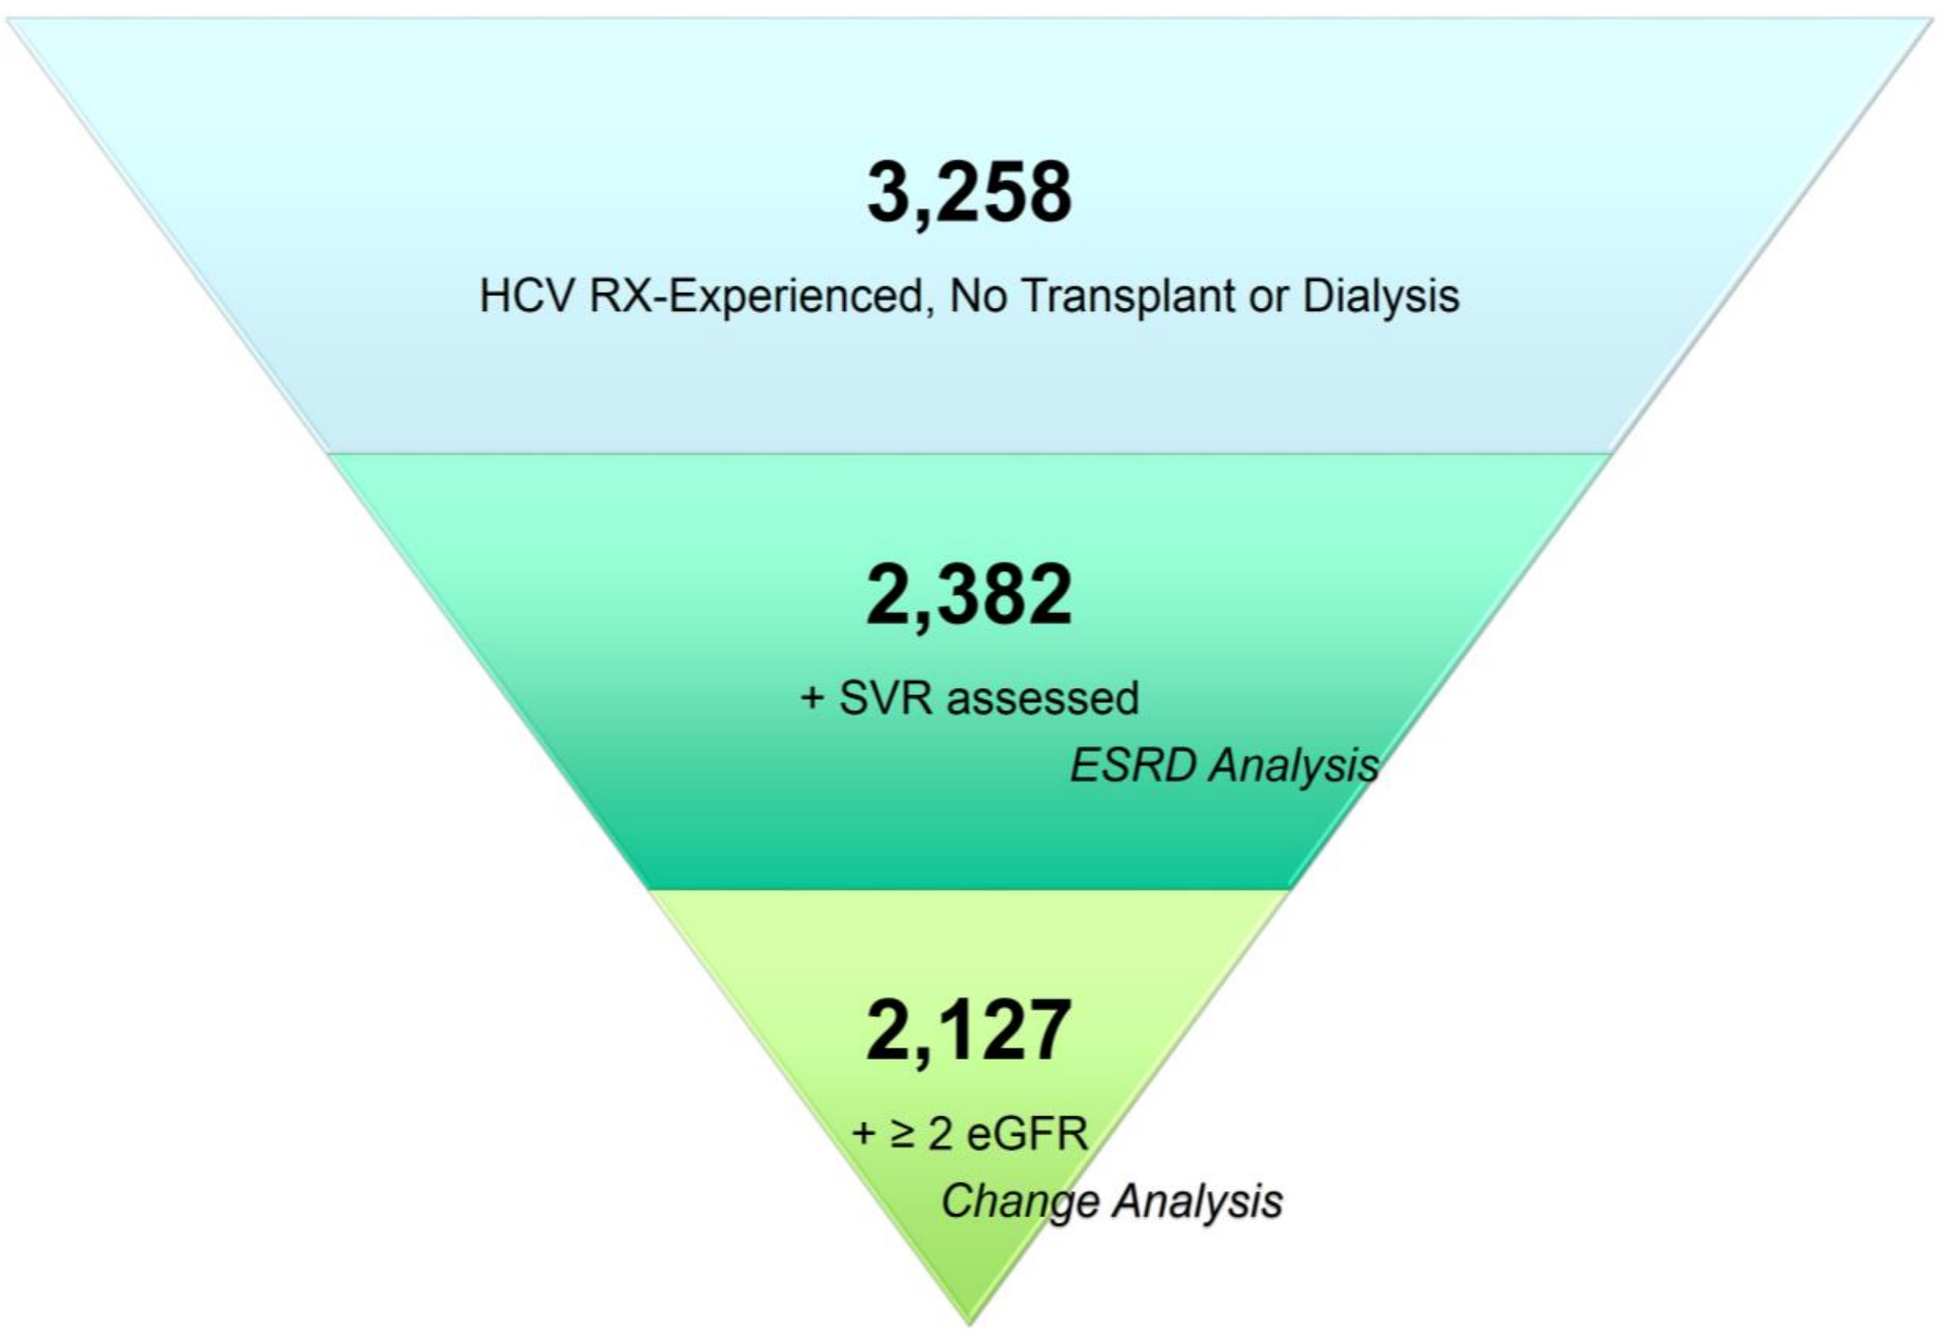
3,258
HCV RX-Experienced, No Transplant or Dialysis
2,382
+ SVR assessed
ESRD Analysis
2,127
+ ≥ 2 eGFR
Change Analysis

Table 1. Clinical and demographic characteristics of HCV patients prior to treatment

	Total (N=2382, 100%)	SVR (n=1499, 62.93%)	No-SVR (n=883, 37.07%)
Age in years, mean (SD)	51.67 (8.99)	50.73 (9.12)	53.26 (8.53)
Sex (female)	930 (39.04)	588 (39.23)	342 (38.73)
Race/Ethnicity			
Asian/Pacific Islander	148 (6.21)	114 (7.61)	34 (3.85)
Black	366 (15.37)	154 (10.27)	212 (24.01)
Hispanic	669 (28.09)	408 (27.22)	261 (29.56)
Multiple/Other	22 (0.92)	18 (1.20)	4 (0.45)
White	1140 (47.86)	780 (52.03)	360 (40.77)
Unknown	37 (1.55)	25 (1.67)	12 (1.36)
Comorbid Conditions			
Cirrhosis	620 (26.03)	298 (19.88)	322 (36.47)
Diabetes mellitus	309 (12.97)	153 (10.21)	156 (17.67)
HIV	51 (2.14)	34 (2.27)	17 (1.93)
HBV	25 (1.05)	20 (1.33)	5 (0.57)
HCV Viral Load			
Reactive	247 (10.37)	163 (10.87)	84 (9.51)
<100,000 IU/mL	274 (11.50)	225 (15.01)	49 (5.55)
≥100,000 IU/mL	1802 (75.65)	1076 (71.78)	726 (82.22)
Baseline eGFR, mean (SD)	91.54 (17.56)	91.63 (16.59)	91.38 (19.10)
HCV Genotype			
1	1280 (53.74)	631 (42.09)	649 (73.50)
2	464 (19.48)	397 (26.48)	67 (7.59)
3	317 (13.31)	237 (15.81)	80 (9.06)
4	50 (2.10)	28 (1.87)	22 (2.49)
5	1 (0.04)	1 (0.07)	0 (0.00)
6	3 (0.13)	0 (0.00)	3 (0.34)

Figure 2. Adjusted annual rate of change in post-RX eGFR by SVR status (n=2,127)

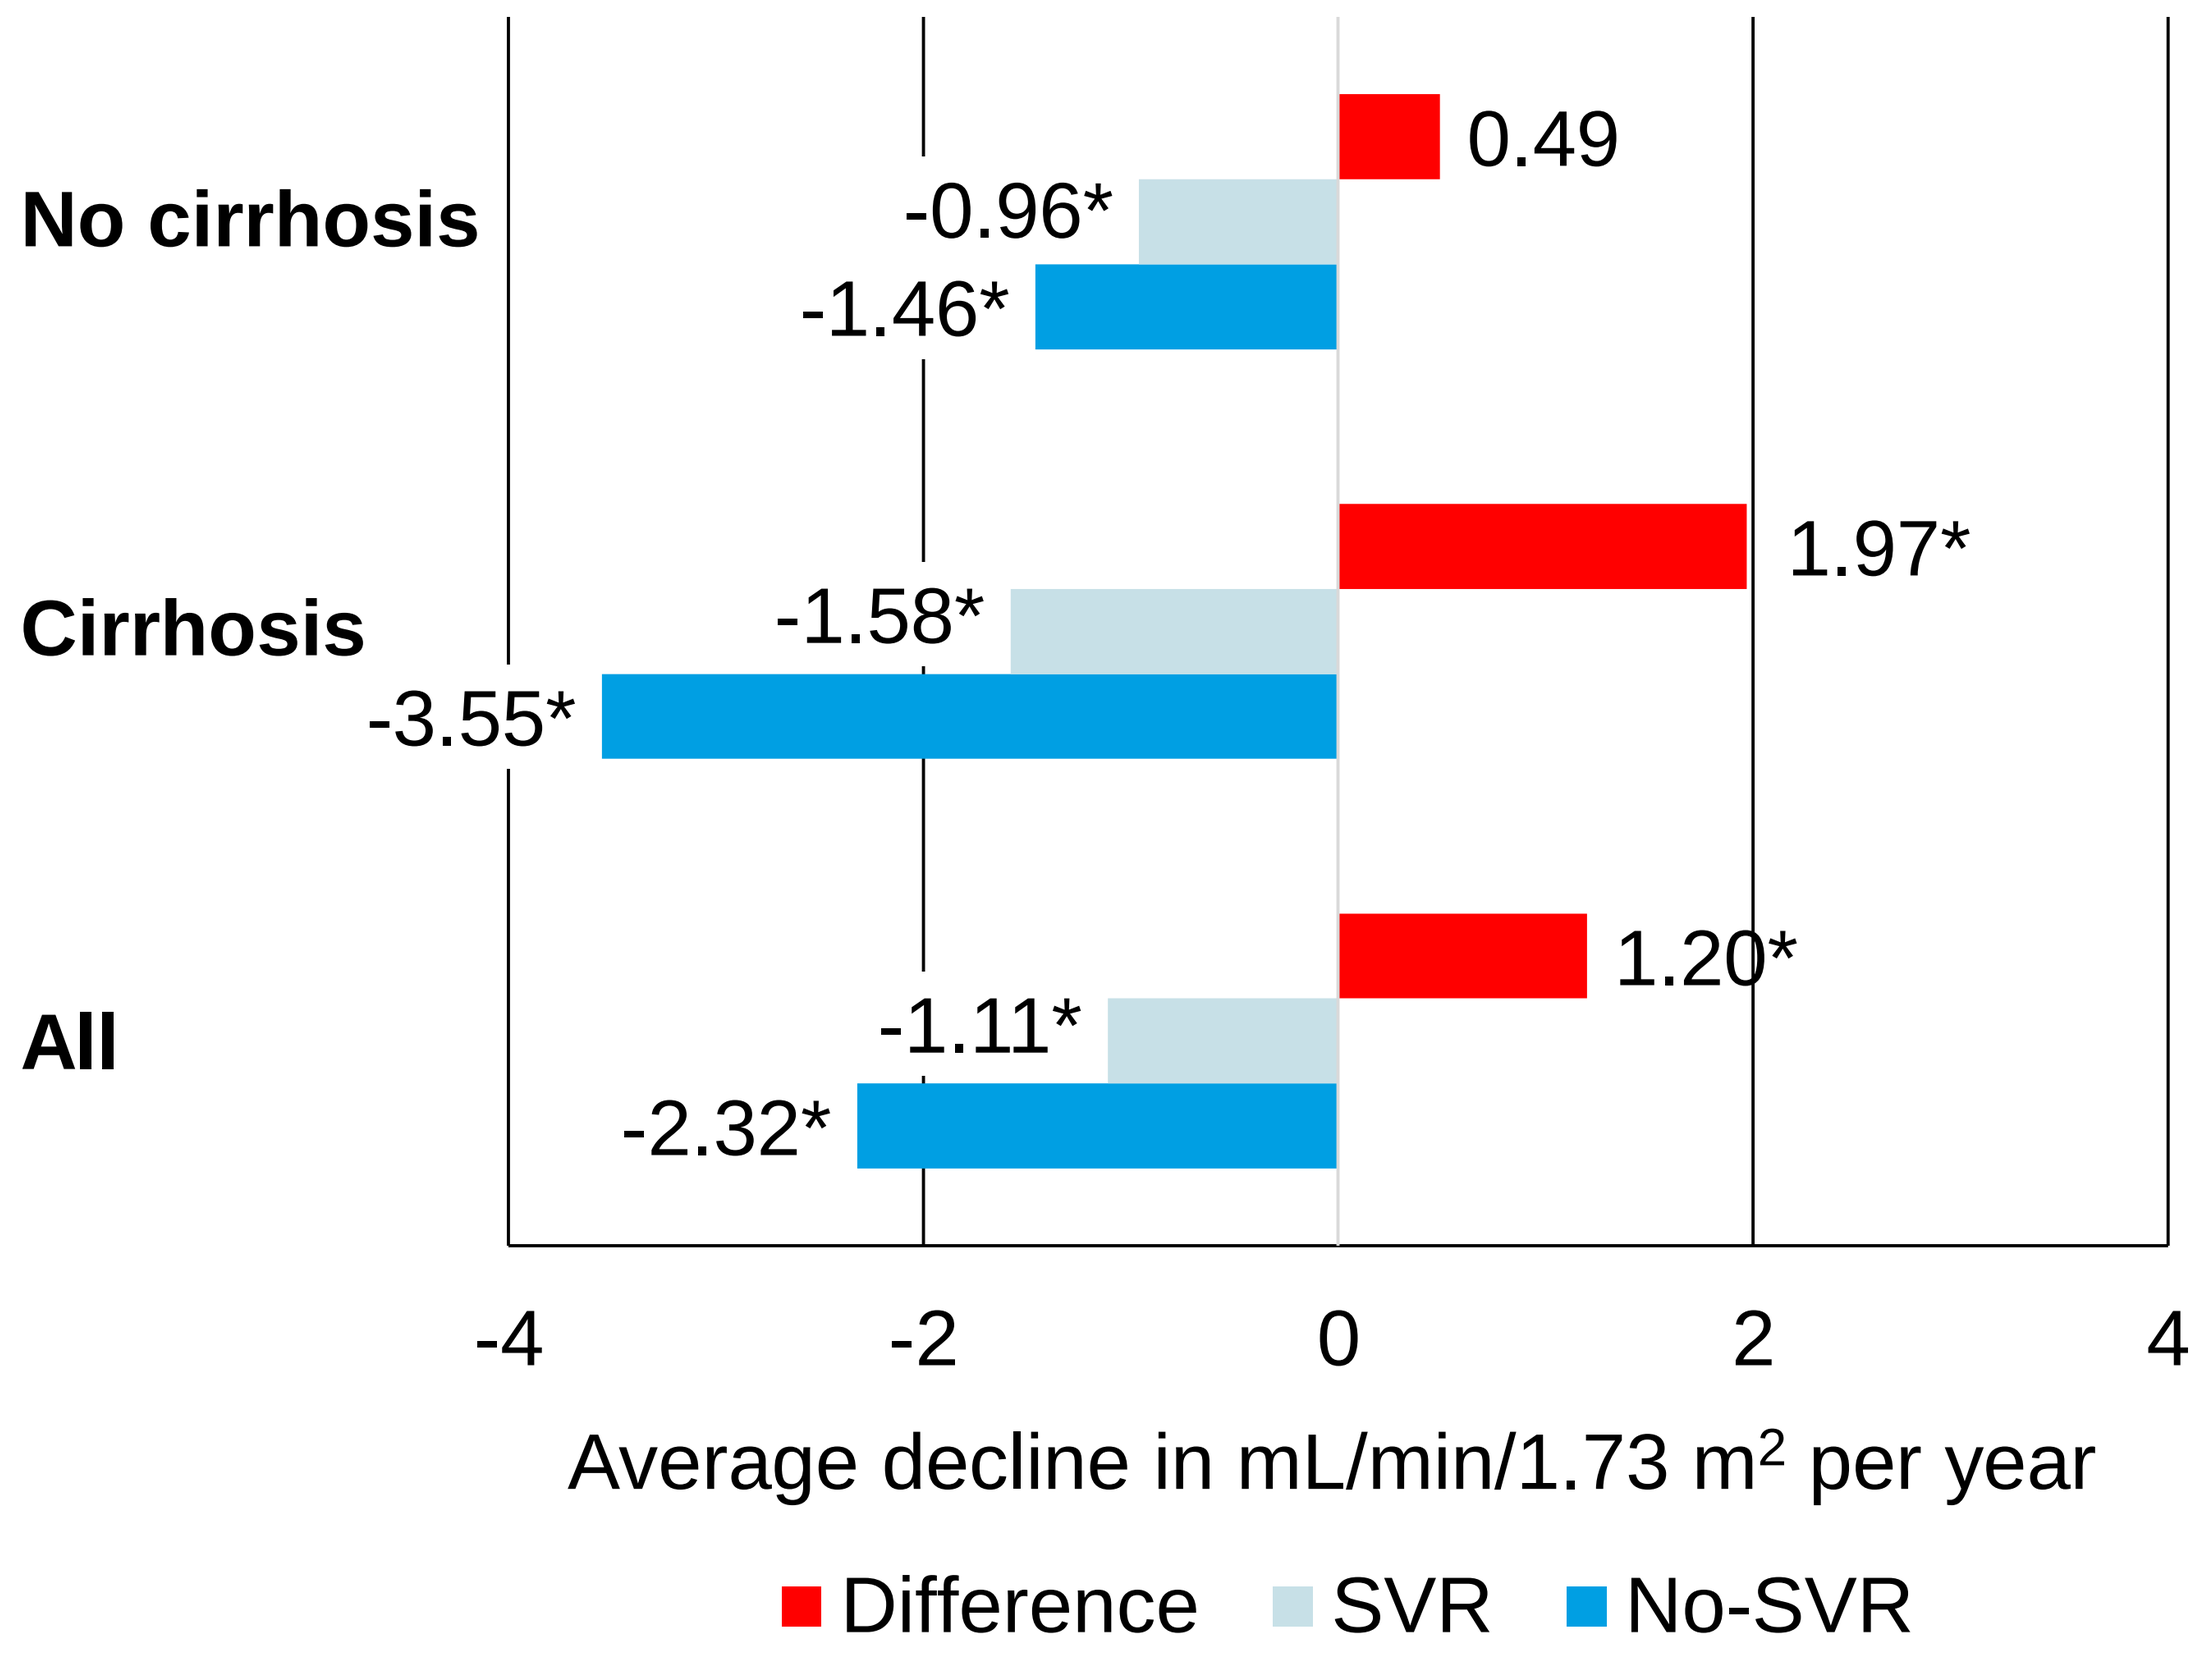
No cirrhosis
0.49
-0.96*
-1.46*
Cirrhosis
1.97*
-1.58*
-3.55*
All
1.20*
-1.11*
-2.32*
-4
-2
0
2
4
Average decline in mL/min/1.73 m2 per year
Difference
SVR
No-SVR

*Statistically significant at *P*<0.05

Table 2. Adjusted hazard ratios for ESRD and 25% decline in eGFR by SVR status

	All	Cirrhosis	No Cirrhosis
ESRD (n=2,382)	0.13 (0.03, 0.56)	0.21 (0.03, 1.40)	No Estimate*
25% Decline (n=2,127)	0.85 (0.70, 1.05)	0.53 (0.37, 0.77)	1.14 (0.88, 1.48)

*No estimate because 0 events observed in SVR group with no cirrhosis in adjusted analysis.

Table 3. Number and percent of patients with 25% eGFR decline or ESRD events by SVR status

		All	Cirrhosis	No Cirrhosis
ESRD	SVR	3 (0.2%)	3 (1.1%)	0 (0.0%)
	No SVR	19 (2.4%)	9 (3.1%)	10 (2.0%)
25% Decline	SVR	275 (24.2%)	69 (27.7%)	206 (23.2%)
	No SVR	247 (32.8%)	119 (41.9%)	128 (27.2%)

- We note differential distribution of genotype, cirrhosis, diabetes, and race by SVR status (**Table 1**)
- The number of eGFR measurements was higher in non-SVR (mean=10.2) vs SVR (mean=6.0)
- The average length of follow-up was ~4 yr in both SVR and non-SVR
- ESRD was a rare event, particularly in those with no cirrhosis and SVR (**Table 3**)

Conclusions

- SVR reduced the average annual rate of renal decline by half (**Figure 2**) and the risk of ESRD by almost 87% (**Tables 2 and 3**)
- The effect of SVR on reducing the annual decline in eGFR (**Figure 2**) and risk of a 25% decline was most pronounced among patients with cirrhosis (**Tables 2 and 3**)
- This study provides additional evidence that achieving cure of HCV infection with an IFN-based therapy results in extrahepatic benefits

Limitations

- Results for the time to ESRD analysis may be unstable due to the small number of observed ESRD events (**Table 3**)
- A larger proportion of Blacks and those with cirrhosis were excluded from analysis due to missing SVR. However, we do not expect that missing Black and cirrhotic patients have worse outcomes than those included in the analysis and thus, do not expect bias from their exclusion
- These results may not be generalizable to cure by IFN-free direct-acting antivirals

KAISER PERMANENTE®